MAJOR HEPATIC RESECTION AS A LIVER GRAFT SAVING PROCEDURE

O. Detry, P. Honoré, B. Detroz, E. Hamoir, T. Defechereux, M. Meurisse, N. Jacquet

Department of Liver Surgery and Transplantation, University Hospital Sart Tilman B35, Liège, Belgium

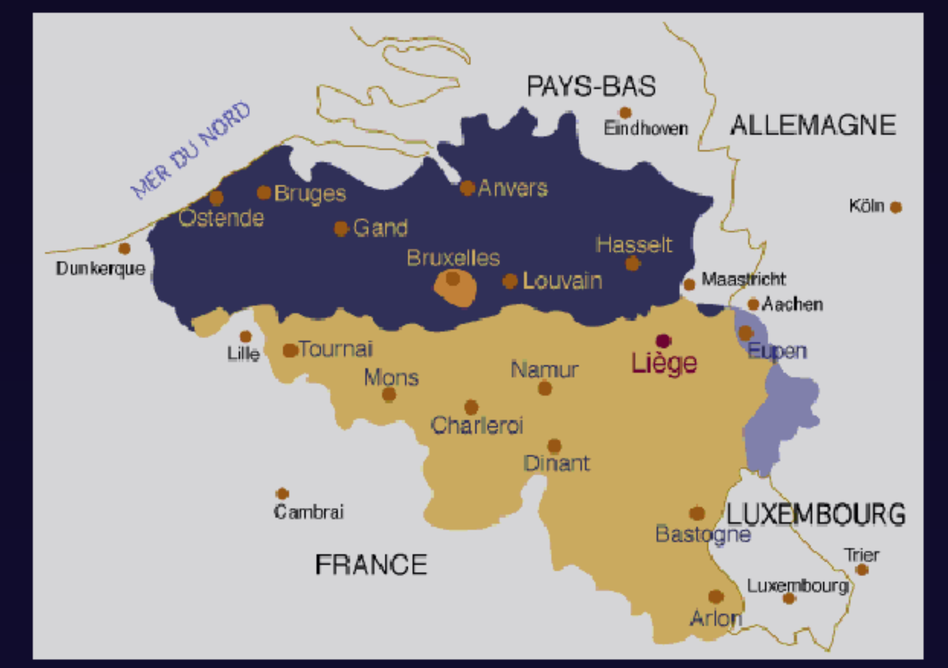
PAYS-BAS
Eindhoven
ALLEMAGNE
MER DU NORD
Bruges
Anvers
Ostende
Gand
Köln
Bruxelles
Hasselt
Dunkerque
Louvain
Maastricht
Aachen
Lille
Tournai
Liège
Eupen
Mons
Namur
Charleroi
Dinant
Cambrai
LUXEMBOURG
Bastogne
FRANCE
Trier
Luxembourg
Arlon

Introduction

Retransplantation for failing graft is quite common after orthotopic liver transplantation (OLT). One of the reasons for liver retransplantation may be intrahepatic biliary strictures, associated or not with hepatic artery thrombosis. However, liver retransplantation consumes a second liver graft in an area of organ shortage (1317 patient deaths on the UNOS waiting list in 1998). Moreover, liver retransplantation is associated with higher morbidity and mortality. Therefore, every attempt to save the liver graft should be performed before retransplantation. The aim of this paper is to report our experience with right hepatectomy as a graft-saving procedure in liver recipients.

Material and Methods

In a consecutive series of 170 adult OLT, 4 male patients (mean age: 45 years, 23 to 55) underwent right hepatectomy (Couinaud's segments V, VI, VII, VIII) in the post transplant period. Causes of liver failure leading to OLT were HCV in 3 cases and chronic liver failure secondary to Wilson's disease in one.
Three patients (Patients 1, 2, 3) developed intrahepatic biliary strictures, related to late hepatic artery thrombosis in one case. As these biliary strictures were mainly localized in the right part of the liver, classical right hepatectomy was performed (delay between OLT and right hepatectomy: 14, 75, 78 months). The fourth patient (Patient 4) developed post transplant ischemic parenchymal infarcts in the right liver. This patient underwent right hepatectomy 48 hours after OLT.

In all cases, classical right hepatectomy was performed without Pringle's maneuver and without mobilization of the left liver and the surrounding adhesions in order to preserve its collateral vascularization. Parenchymal transection was performed with an ultrasonic dissector and with the use of clips for hemostasis and biliostasis. The transected surface was then sprayed with fibrin glue. A cell saver was used in all procedures.

Patient 1 was born in 1967. In 1987, he underwent OLT for decompensated cirrhosis secondary to Wilson's disease. He developed intrahepatic bile duct strictures secondary to hepatic artery thrombosis and was retransplanted in 1989. This second liver transplantation was complicated by hepatic artery thrombosis causing extra- and intra- hepatic liver strictures. In 1991, he underwent bile duct derivation with a Roux-in-Y choledocojejunostomy, and hepatic artery revascularization using in situ fibrinolysis and termino-terminal anastomosis between the right gastroepiploic and the main hepatic arteries. The left branch of the hepatic artery was successfully repermeabilized, but the right artery was lost.

The patient suffered from recurrent episodes of cholangitis. As the strictures were mainly in the right bile duct branch (Fig 1.1), the patient underwent right hepatectomy in 1996. At follow-up in January 2000, the patient was asymptomatic. He has not experienced cholangitis since the hepatectomy. Ultrasonography and doppler assessed the permeability of the hepatic artery and the compensatory hypertrophy of the remnant left liver (Fig 1.2).

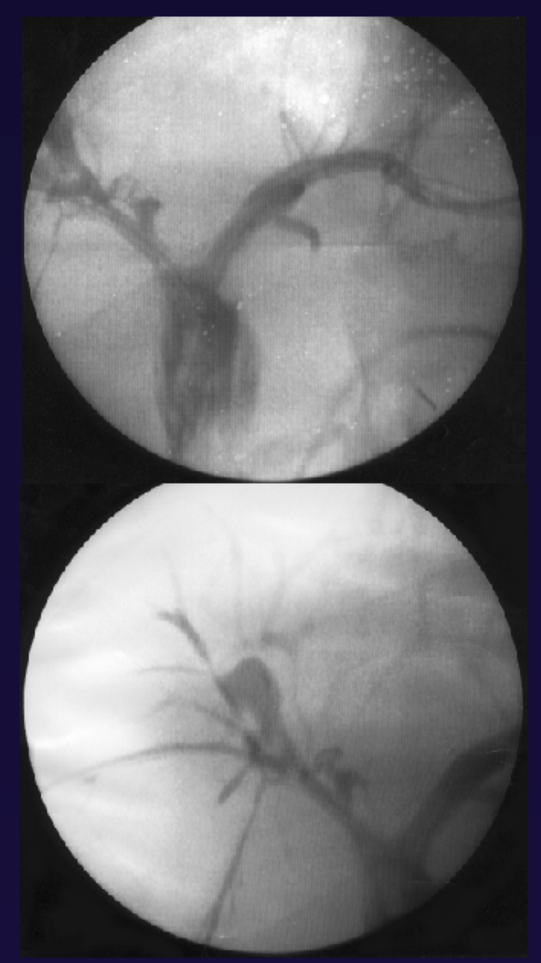
Fig 1.1

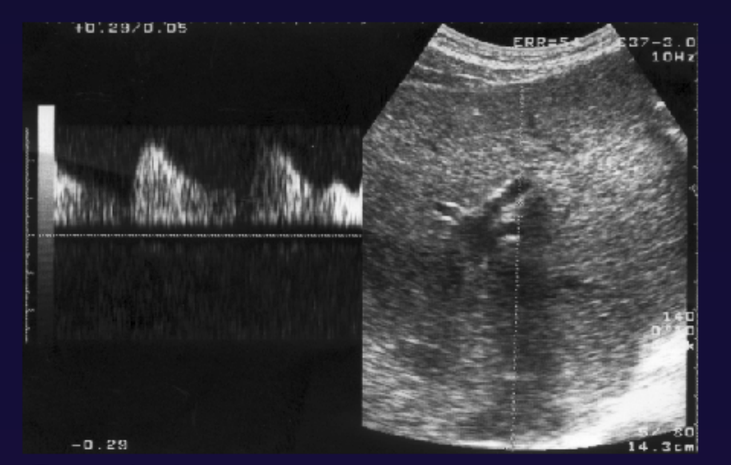
Fig 1.2

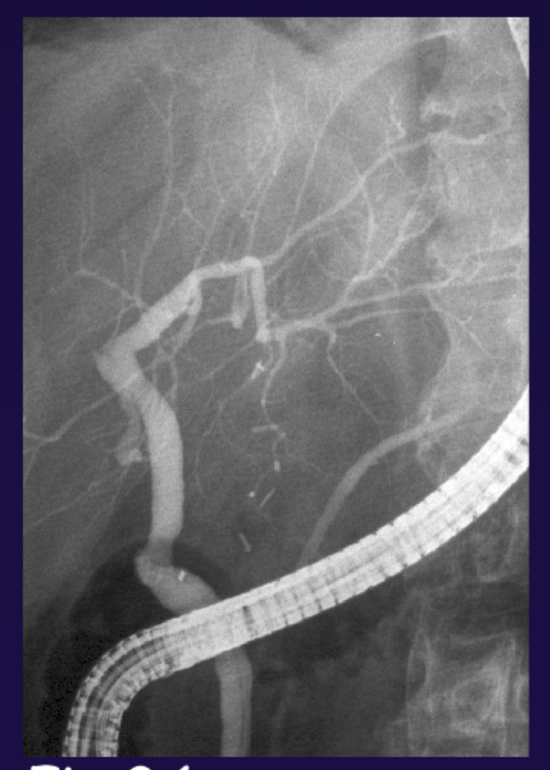
Fig 2.1

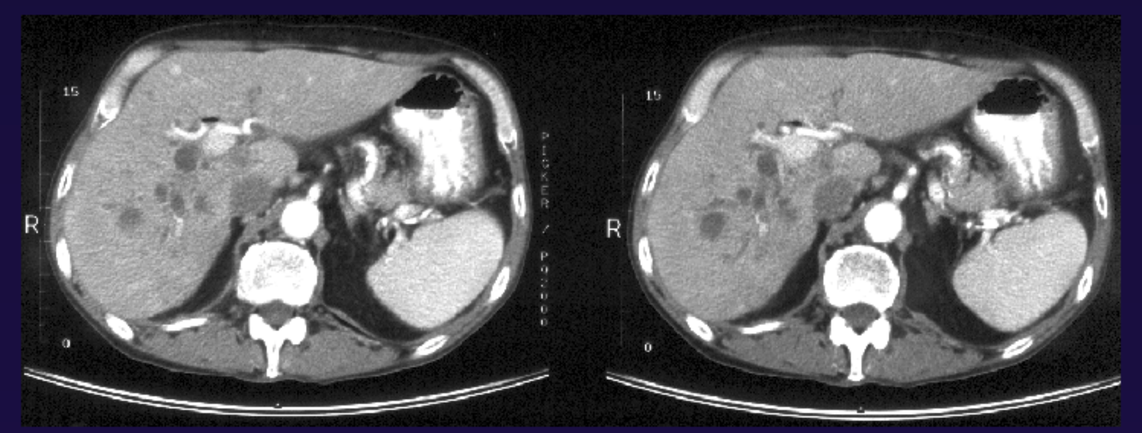
Fig 2.2

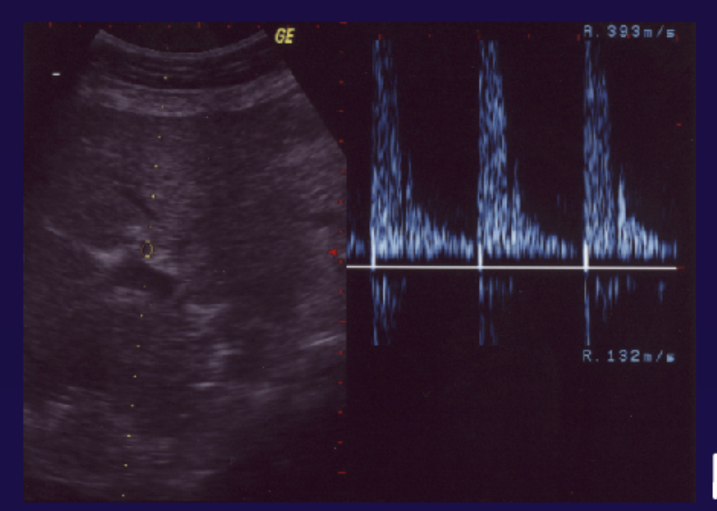
Fig 2.3

Patient 2 is a man born in 1935. He underwent OLT in 1990 for decompensated cirrhosis secondary to HCV. In 1996, he underwent endoscopic retrograde cholangiography for recurrent cholangitis, that revealed right bile duct branch stenosis. Hepatic artery was permeable as assessed by doppler ultrasonography. Endoscopic treatment with stents was not successful and the right bile duct branch was considered as occluded (Fig 2.1). In 1996 he required hemodialysis for chronic renal insufficiency, in part related to cyclosporin. In September 1996 he was hospitalized for recurrent sepsis and abdominal computed tomography (CT) revealed abcesses in the right liver (Fig 2.2). He underwent right hepatectomy in October 1996 without complication, and kidney transplantation in January 1997. At follow-up in April 2000, the patient was asymptomatic. He has not experienced infection since the hepatectomy. Ultrasonography and doppler assessed the permeability of the hepatic artery (Fig 2.3) and compensatory hypertrophy of the remnant left liver.

Patient 3, a male patient born in 1947, underwent uncomplicated OLT in August 1997 for decompensated HCV cirrhosis. In October, he was readmitted for cholangitis. Hepatic artery was permeable. Retrograde cholangiography showed strictures mainly in the intrahepatic right bile duct branch (Fig 3.1). This stricture was treated with a stent. In a few months, he developed five recurrences of cholangitis requiring hospitalizations and several cholangiographies confirmed the intrahepatic bile duct strictures localized in the right liver. Endoscopic treatments with stents and balloons were unsuccessful; in September 98 a CT scanner of the liver showed hypovascularized regions of the right liver, attributed to local infection (Fig 3.2). In October 1998 the patient underwent right hepatectomy. During the procedure, abscesses of the right liver were demonstrated. The operation was complicated by a biliary fistula treated conservatively. Since the hepatectomy the patient was not readmitted for cholangitis. An abdominal CT scanner was performed for exploration of aspecific abdominal pain. This scanner showed compensatory hypertrophy of the left remnant liver (Fig 3.3).

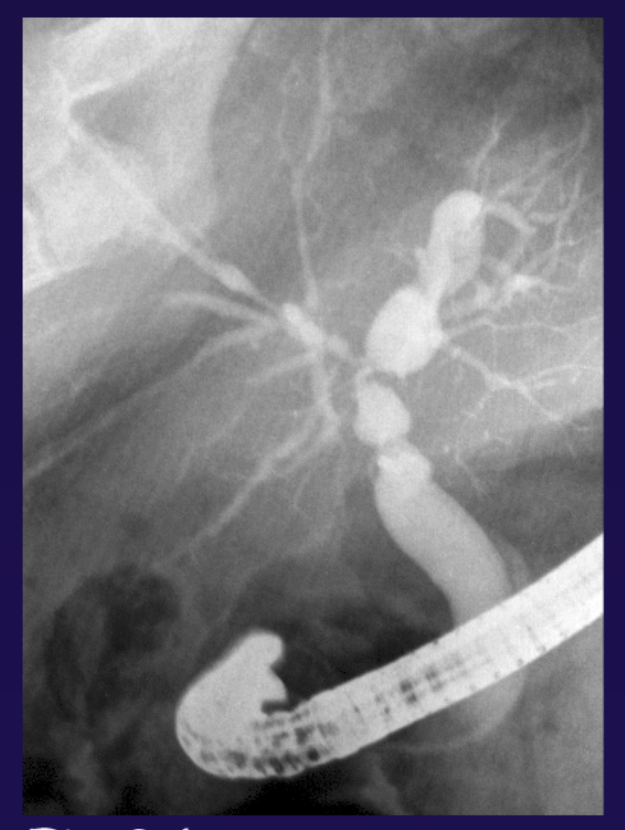
Fig 3.1

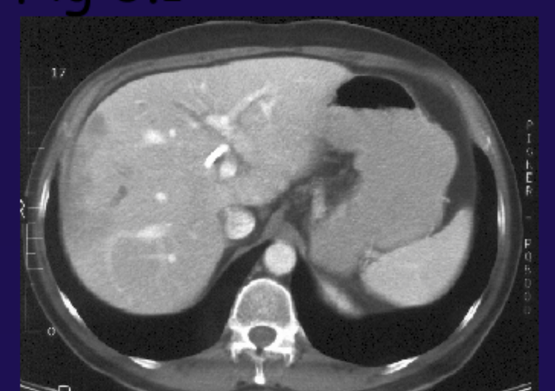
Fig 3.2

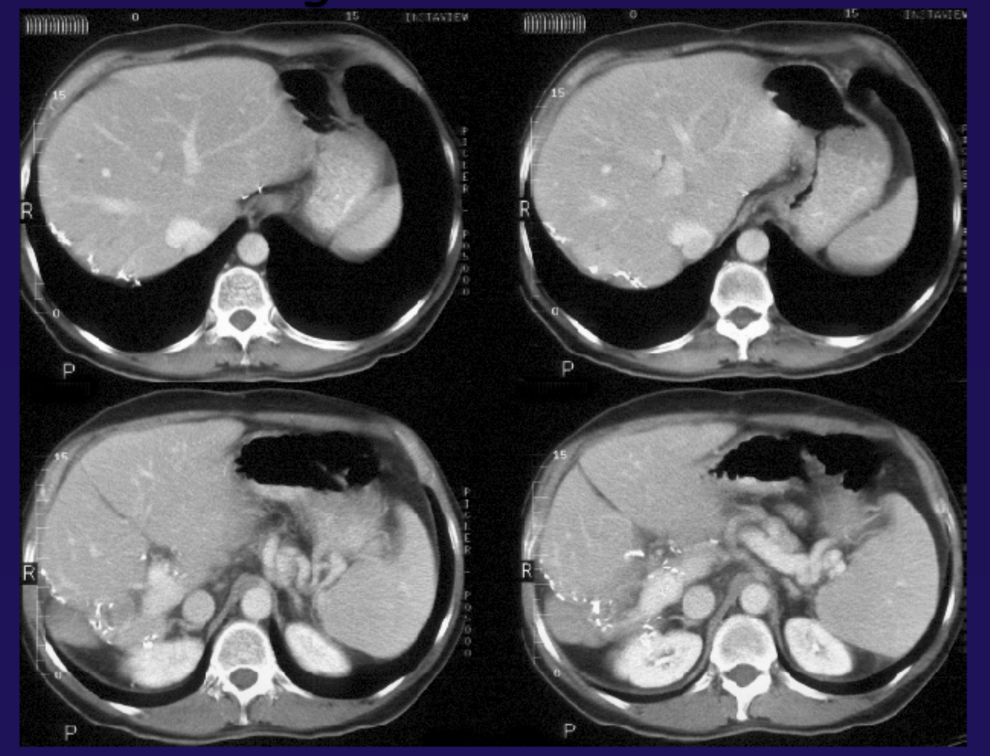
Fig 3.3

Patient 4 was born in 1945. He developed HCV cirrhosis complicated by a 6 cm hepatocarcinoma treated by chemoembolization in December 1996 and segment VII resection in January 1997 (Fig 4.1). In November 1998, he underwent OLT for decompensated cirrhosis, with a Roux-in-Y choledocojejunostomy. At the second postoperative day the patient developed major cytolysis and abdominal CT scanner showed right liver necrosis (Fig 4.2). At laparotomy the necrosis of the right liver was confirmed and right laparotomy was performed. The hepatic artery was permeable. The patient left the hospital at postoperative day 22 without complication. At the third month follow-up the hepatic artery was not found. Hepatic artery thrombosis was confirmed by selective arteriography. He developed progressive icterus and prurit secondary to ischemic cholangitis and he was placed on the waiting list for retransplantation in April 2000.

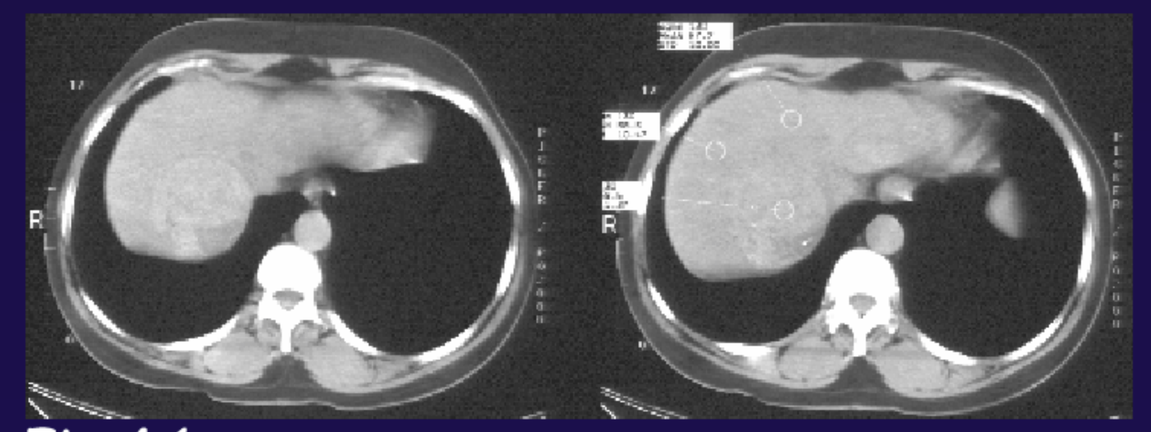
Fig 4.1

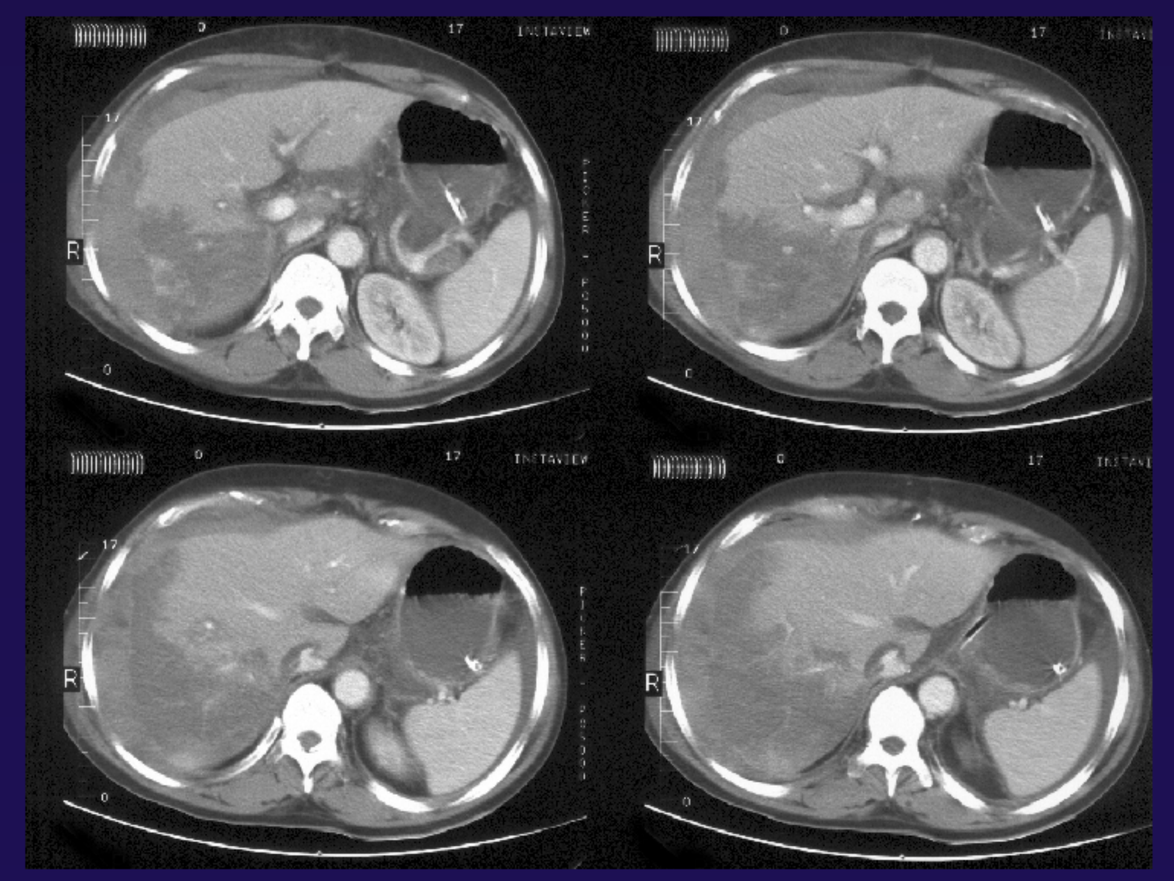
Fig 4.2

Results

All patients are alive without retransplantation (mean follow-up 28 months, 13 to 48). One patient developed postoperative biliary fistula treated conservatively. Mean postoperative hospital stay was 20 days.
Patient 4 developed significant cholostasis secondary to late artery thrombosis and was replaced on the waiting list for retransplantation.

Conclusions

Major hepatectomy may be proposed as graft saving procedure in selected liver recipients with good outcome on both patient and graft survival. The main indication seems to be preservation injury to the intrahepatic bile ducts that can be sometimes localized to a liver lobe. In our experience, diffuse intrahepatic bile duct strictures are difficult to manage conservatively and often require retransplantation.